Sex-specific differences in bone mass are maintained following spinal cord injury

Courtney M. Mazur[1], W. Brent Edwards, Ifaz T. Haider, Ying Fang, Leslie R. Morse, Thomas J. Schnitzer, Narina Simonian, & Karen L. Troy[2]

[2]Department of Biomedical Engineering, Worcester Polytechnic Institute, Worcester, MA

Email: [1]CMMAZUR@mgh.harvard.edu, [2]ktroy@wpi.edu

INTRODUCTION

Spinal cord injuries (SCI) cause rapid, severe bone loss in the distal femur and proximal tibia and increase risk of low-impact fracture[1]. The rate of bone loss and the affected sites are distinct from osteoporosis following menopause or aging, so SCI-induced bone loss requires specific attention. While women are at higher risk of osteoporosis than men, individual studies of SCI-induced bone loss typically do not enroll enough women to discern sex-specific patterns following SCI.

Objective:
This investigation analyzed QCT scans from four clinical trials with nearly identical imaging protocols to quantify sex-specific differences in bone mineral content following SCI.

METHODS

Subjects

- 137 adults (31 female), 0-50 years post-SCI (injury duration: 8.7±10.4 years, age: 40±15 years, mass: 78±19 kg)
- Baseline QCT scans from and one observational study (PIs: Schnitzer and Troy) and four clinical trials were used (NCT02325414, PI: Schnitzer; NCT01225055, PI: Schnitzer; NCT01426555, PI: Morse; NCT02533713, PI: Morse)
- Protocols were approved by the IRB at each study site, and all participants provided informed consent [2-4].

Data Collection

- 30 cm long scans contained 15 cm each of the distal femur and proximal tibia from one limb per individual.
- A hydroxyapatite calibration phantom was used to convert CT absorption to mineral density for each subject.
- Scans were aligned and segmented to include all tissue inside the periosteal boundary (0.15 g/cm^3 threshold).
- Regions of interest (epiphysis, metaphysis, diaphysis) were assigned as successive 10% bone length sections.

ANALYSIS

- Integral, trabecular, and cortical volumes of the femoral and tibial epiphysis, metaphysis and diaphysis were analyzed.
- For each volume and region, bone parameters were plotted as a function of time post-SCI. Men's and women's data were fit with single-phase exponential decay curves [3,4]
- Later, the acute (<6 months) and plateau (>3 years) phases post-injury were compared with two-way ANOVA.

Term	Definition
Integral	All voxels within periosteal surface
Trabecular	All integral voxels > 3.5 mm from periosteal surface
Cortical	Integral voxels < 3.5 mm from periosteal surface with density > 0.35 g/cm^3
BV	Bone volume, cm^3
BMC	Bone mineral content, g
BMD	Bone mineral density, g/cm^3
BSI	Bending strength index, cm^3 (I_x+I_y / Width)

RESULTS

As in prior analyses [4], we found that integral bone volume (BV) in both bones and all regions does not change after SCI. Female integral BV was on average 70-77% of that in males, depending on region (Fig. 1, p<0.0001).

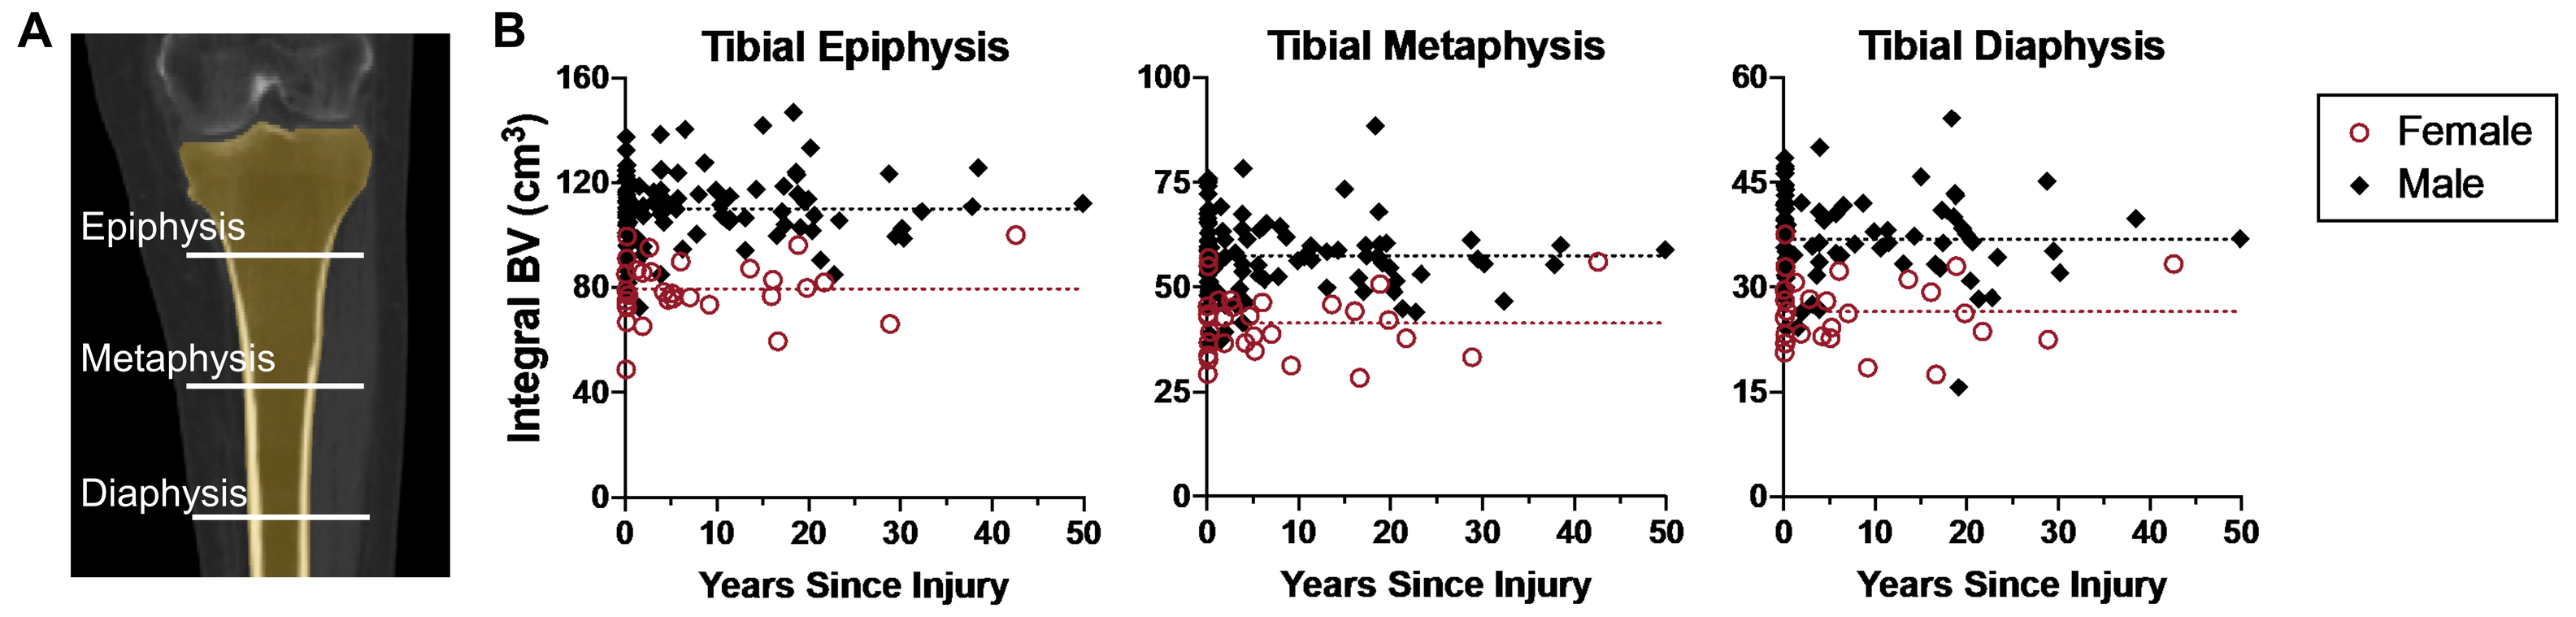
A
Epiphysis
Metaphysis
Diaphysis
B
Tibial Epiphysis
Tibial Metaphysis
Tibial Diaphysis
Integral BV (cm³)
Years Since Injury
Female
Male

Figure 1: A Diagram of regions of the interest in the tibia. **B** Integral BV in the proximal tibia as a function of time since injury. Dashed lines indicate mean BV for each sex.

Bone mineral content (BMC) rapidly declined until reaching a plateau approximately 3 years post-injury. The decline in BMC over time in each region of interest was well described by separate single-phase exponential decay curves for each sex (Fig 2A, p<0.001). However, when BMC was normalized to the originally-calculated y-intercepts, the data were best fit by one curve for both sexes (Fig 2B).

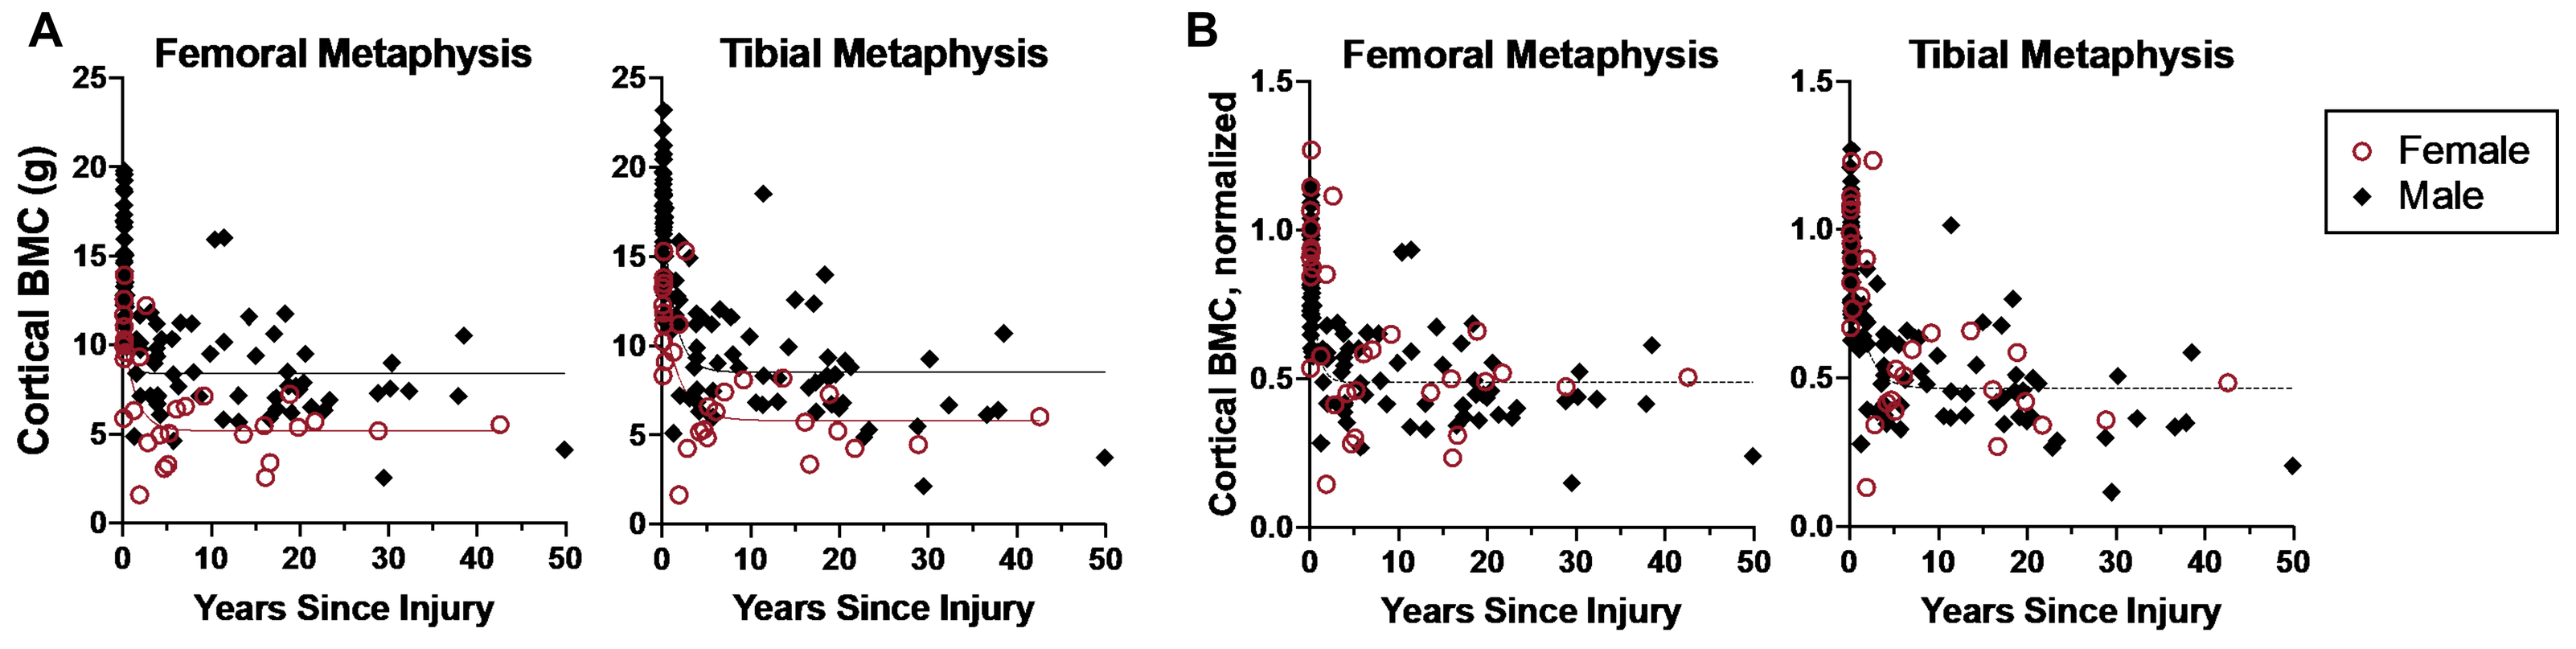
A
Femoral Metaphysis
Tibial Metaphysis
Cortical BMC (g)
B
Femoral Metaphysis
Tibial Metaphysis
Cortical BMC, normalized
Years Since Injury
Female
Male

Figure 2: A Cortical BMC as a function of time post-SCI. **B** Cortical BMC normalized to baseline for each sex. Lines indicate best-fit exponential decay curves (units: g/baseline g from curve fit).

Differences in BMC over time between male and female subjects were due to lower mean values for females in the early (<6 months post-SCI) and plateau (>3 years post-SCI) phases. From the early to the plateau phase, BMC in the epiphysis, metaphysis and diaphysis was reduced by approximately 63%, 54%, and 40%, respectively, for both sexes, with significant differences between the sexes at both times in most regions.

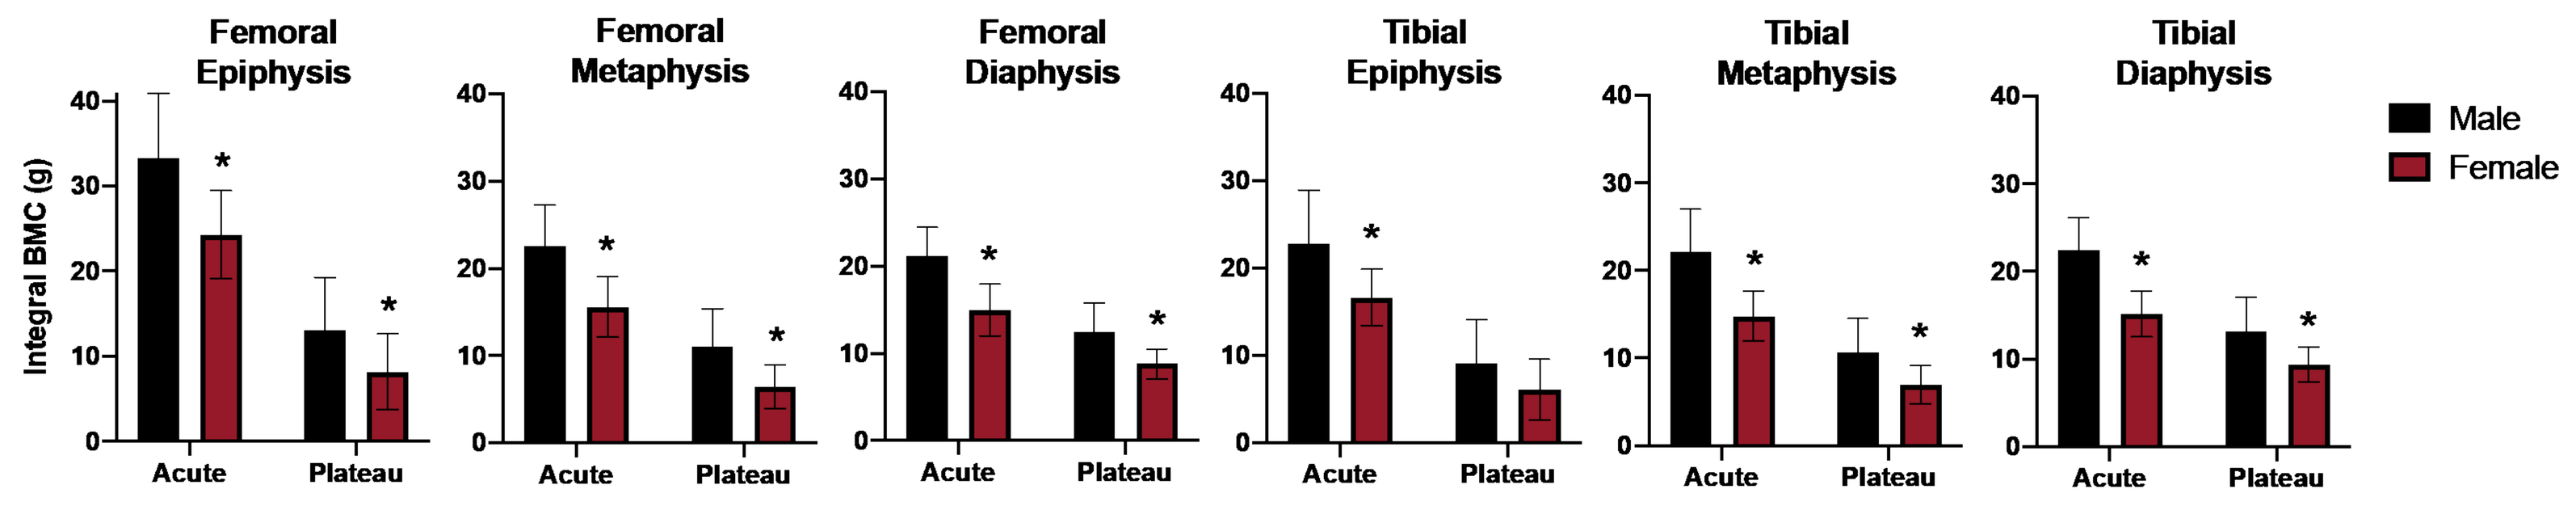
Femoral Epiphysis
Femoral Metaphysis
Femoral Diaphysis
Tibial Epiphysis
Tibial Metaphysis
Tibial Diaphysis
Integral BMC (g)
Acute
Plateau
Male
Female

Figure 3: Integral BMC in females and males in the acute (<6 months) and plateau (>3 years) phases post-injury. *p<0.05 by two-way ANOVA with Tukey's multiple comparisons. N=10-54.

BMD also declined over time post-SCI in all regions. The consistent effect of female sex on BV and BMC leads to equivalent BMD over time in the epiphysis and metaphysis. BMD in the diaphysis was best described by separate decay curves for each sex (p=0.01), due to significantly different rate constants.

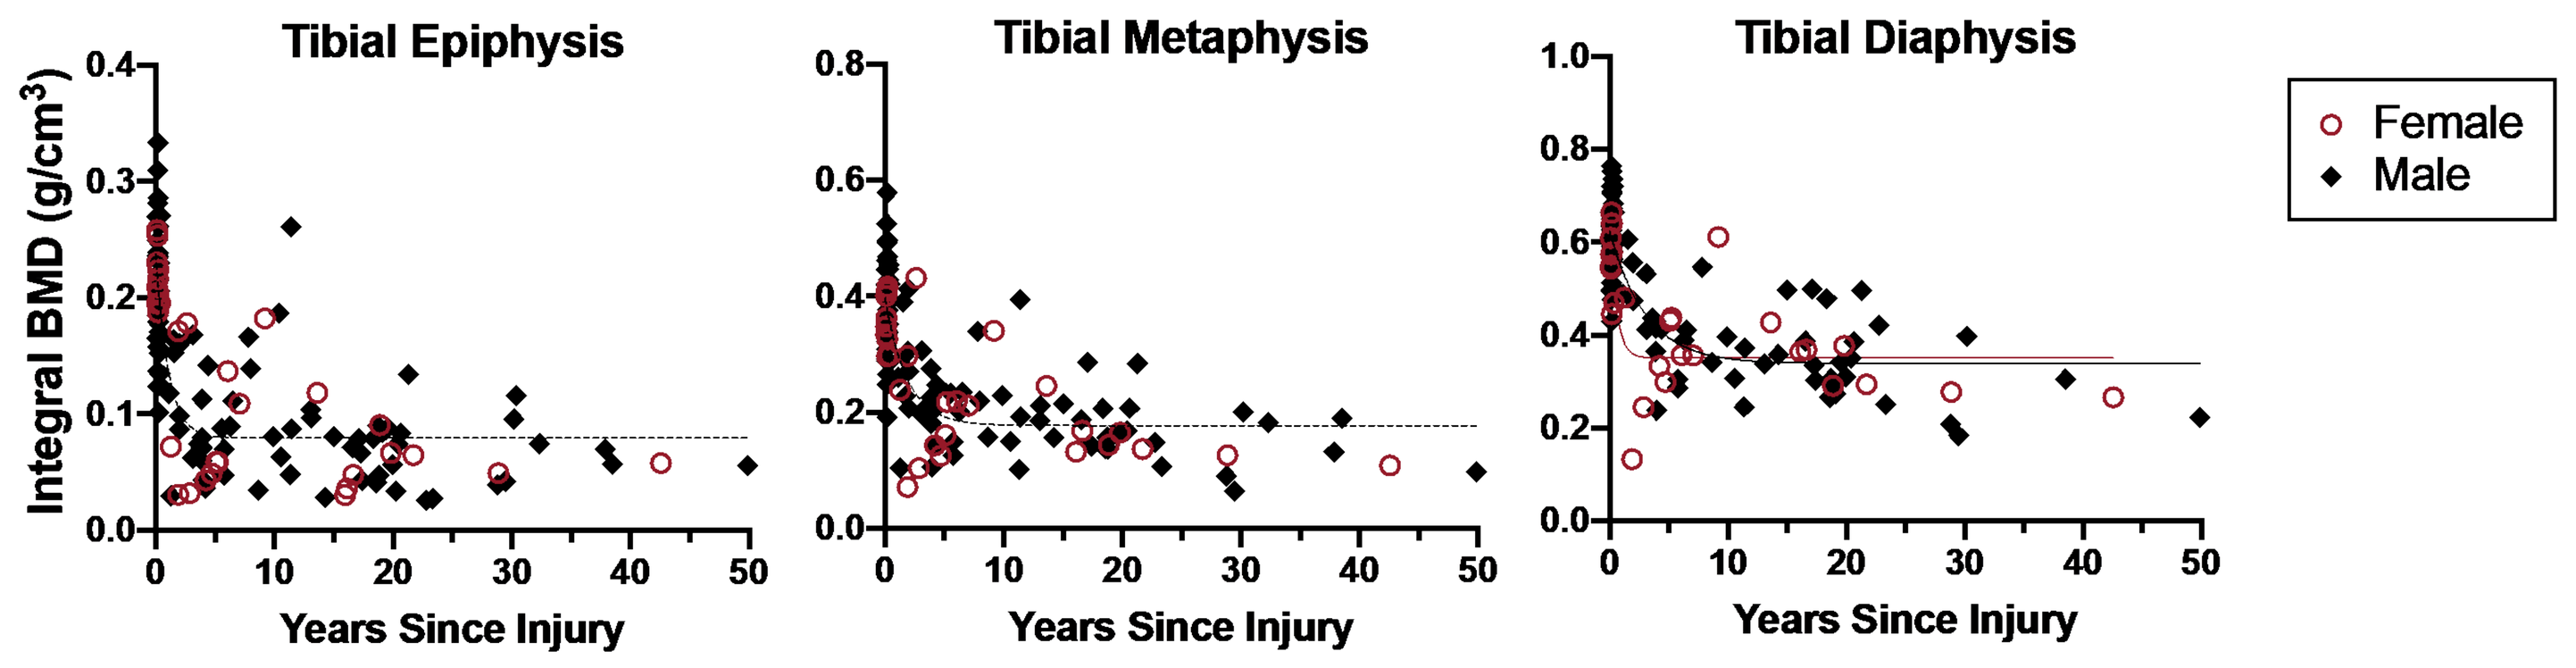
Tibial Epiphysis
Tibial Metaphysis
Tibial Diaphysis
Integral BMD (g/cm³)
Years Since Injury
Female
Male

Figure 4: Integral BMD as a function of time post-injury. Lines indicate best-fit exponential decay curves.

As expected with lower BV and BMC, the bending strength index (BSI) in female bones was significantly different than in males over time (p<0.001), with lower BSI in female bones extending into the plateau phase.

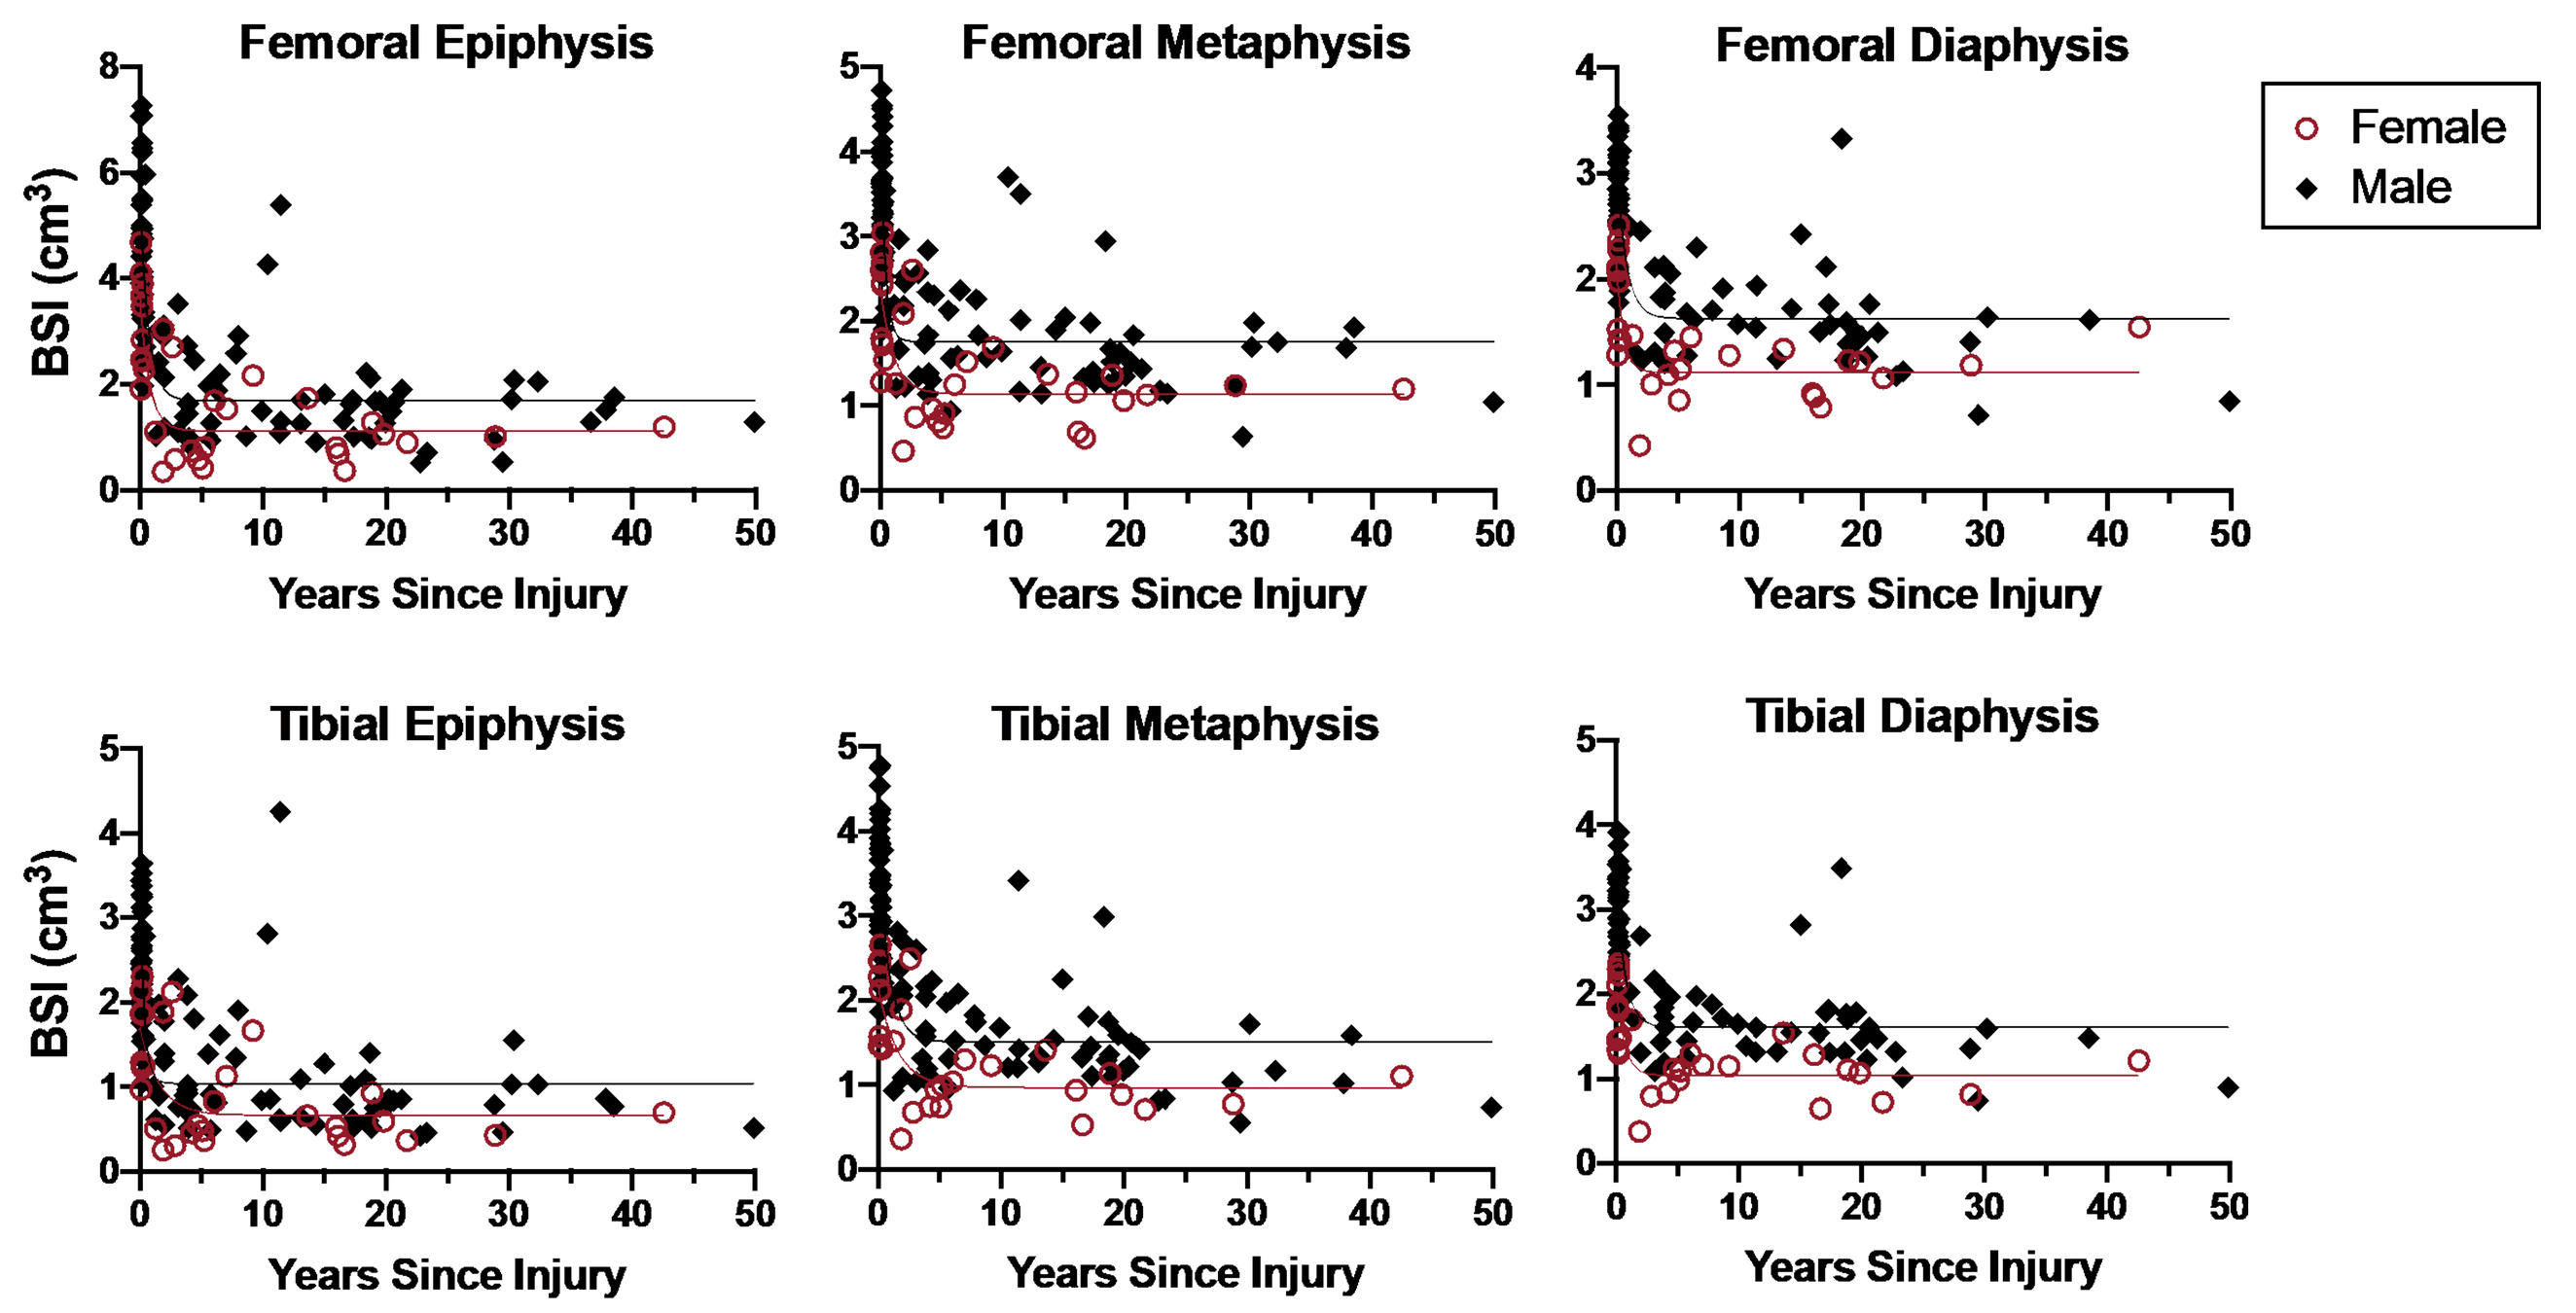
Femoral Epiphysis
Femoral Metaphysis
Femoral Diaphysis
Tibial Epiphysis
Tibial Metaphysis
Tibial Diaphysis
BSI (cm³)
Years Since Injury
Female
Male

Figure 5: BSI in the femur and tibia as a function of time post-SCI. Lines indicate best-fit exponential decay curves.

DISCUSSION

- By combining baseline data from multiple studies, we have the statistical power to determine sex-specific differences in loss of bone mineral over time post-SCI.
- Together these data demonstrate that women have lower BV, BMC, and BSI than men in the acute and plateau phases following SCI.
- The magnitude of SCI-induced bone loss is similar in males and females. Additional data from females 0.5-3 years post-SCI could help to determine whether differences in the rate of bone loss exist.
- Our conclusions are limited by the cross-sectional design of this study. The study design could also be strengthened by the inclusion of a non-injured reference group.
- Due to consistently lower BV, BMC, and BSI, special attention should be paid to female SCI patients to prevent low-impact fractures.

ACKNOWLEDGEMENTS

The data analyzed here were collected with funding from: Department of Defense U.S. Army Medical Research and Materiel Command (Grant Numbers SC090010 , SC130125, W81XWH-10-1-1043 and W81XWH-15-2-0078) and by an investigator-initiated grant from Merck, Inc.

REFERENCES

[1] Frotzler et al. Spinal Cord. 2020 Apr;58(4):441-448.
[2] Morse et al. JBMR Plus. 2019 May; 3(5): e10167.
[3] Haider et al. Osteoporos Int. 2018 Dec; 29(12): 2703–2715.
[4] Edwards et al. J Bone Miner Res. 2015 Aug;30(8):1422-30.